Zenocutuzumab efficacy and safety in advanced *NRG1+* cholangiocarcinoma: Analysis from the phase 2 eNRGy trial

Alison M Schram,[1] James M Cleary,[2] Dirk Arnold,[3] Antoine Hollebecque,[4] Olumide Gbolahan,[5] Misako Nagasaka,[6] Salvatore Siena,[7] Frans Opdam,[8] Yu Sunakawa,[9] Philippe A Cassier,[10] Gaurav Trikha,[11] Vaia Florou,[12] Richard Greil,[13] Paul F La Porte,[14] Pritesh Gandhi,[15] Fiona Garner,[15] Ernesto Wasserman,[16] Shola Adeyemi,[16] Christoph Springfeld[17]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Dana-Farber Cancer Institute, Boston, MA, USA; [3]Asklepios Klinik Altona, Hamburg, Germany; [4]Gustave Roussy, Villejuif, France; [5]Emory Winship Cancer Institute, Atlanta, GA, USA; [6]University of California Irvine Health Chao Family Comprehensive Cancer Center, Orange, CA, USA; [7]Asst Grande Ospedale Metropolitano Niguarda Niguarda Cancer Centre, Milano, Italy; [8]Netherlands Cancer Institute, Amsterdam, Netherlands; [9]St. Marianna University Hospital, Kawasaki-Shi, Japan; [10]Centre Léon Bérard, Lyon, France; [11]Cancer Specialists of North Florida, Jacksonville, FL, USA; [12]Huntsman Cancer Institute, Salt Lake City, UT, USA; [13]Salzburger Universitätsklinikum, Salzburg, Austria; [14]The Oncology Institute of Hope and Innovation - Long Beach, Cerritos, CA, USA; [15]Partner Therapeutics, Inc., Lexington, MA, USA; [16]Merus N.V., Cambridge, MA, USA; [17]Heidelberg University Hospital, Heidelberg, Germany

Background

- Cholangiocarcinoma is a rare, aggressive gastrointestinal cancer with a poor prognosis (median survival in advanced disease of ~1 year)[1–4]
- *NRG1* gene fusions are rare oncogenic drivers (prevalence <1% in cholangiocarcinoma) associated with recurrence, metastases, chemoresistance, and poor prognosis in solid tumors[5–7]
- No approved targeted therapies for *NRG1+* cholangiocarcinoma exist; cholangiocarcinoma treatment is limited to palliative systemic therapy for metastatic disease[1]
- BIZENGRI® (zenocutuzumab) received accelerated US FDA approval (December 2024) for previously treated, advanced *NRG1+* non-small cell lung cancer (NSCLC) and pancreatic adenocarcinoma (PDAC)[8]
- Zenocutuzumab is a HER2/HER3 common light chain bispecific antibody that blocks HER3-mediated NRG1 signaling and HER2/HER3 dimerization and mediates antibody-dependent cellular cytotoxicity (Figure 1)

Figure 1 | Zenocutuzumab mechanism of action

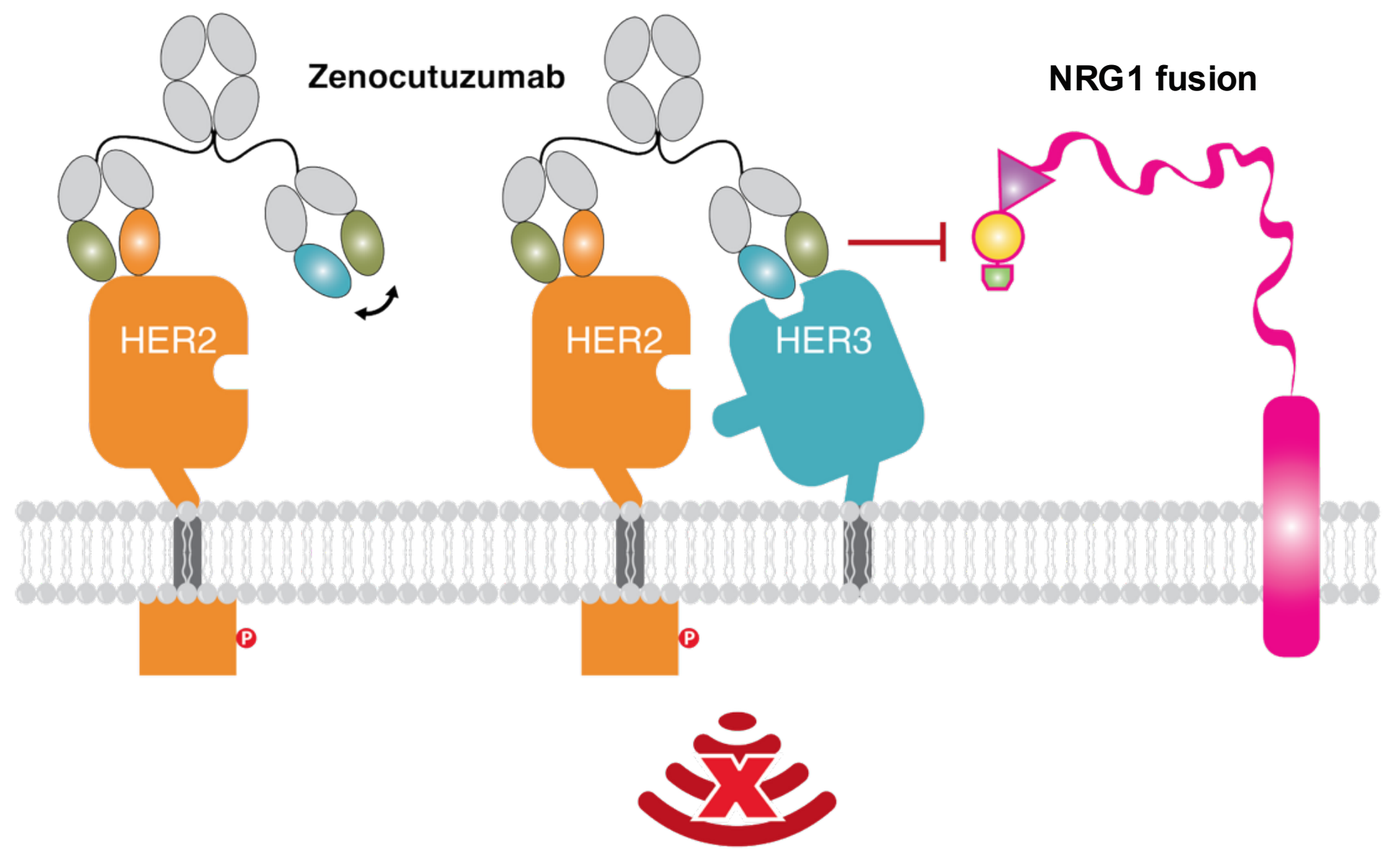
Zenocutuzumab
NRG1 fusion
HER2
HER2
HER3

HER, human epidermal growth factor receptor; NRG1, neuregulin 1.

Methods

Figure 2 | Phase 1/2, global, multicenter zenocutuzumab trial (NCT02912949)[9,10]

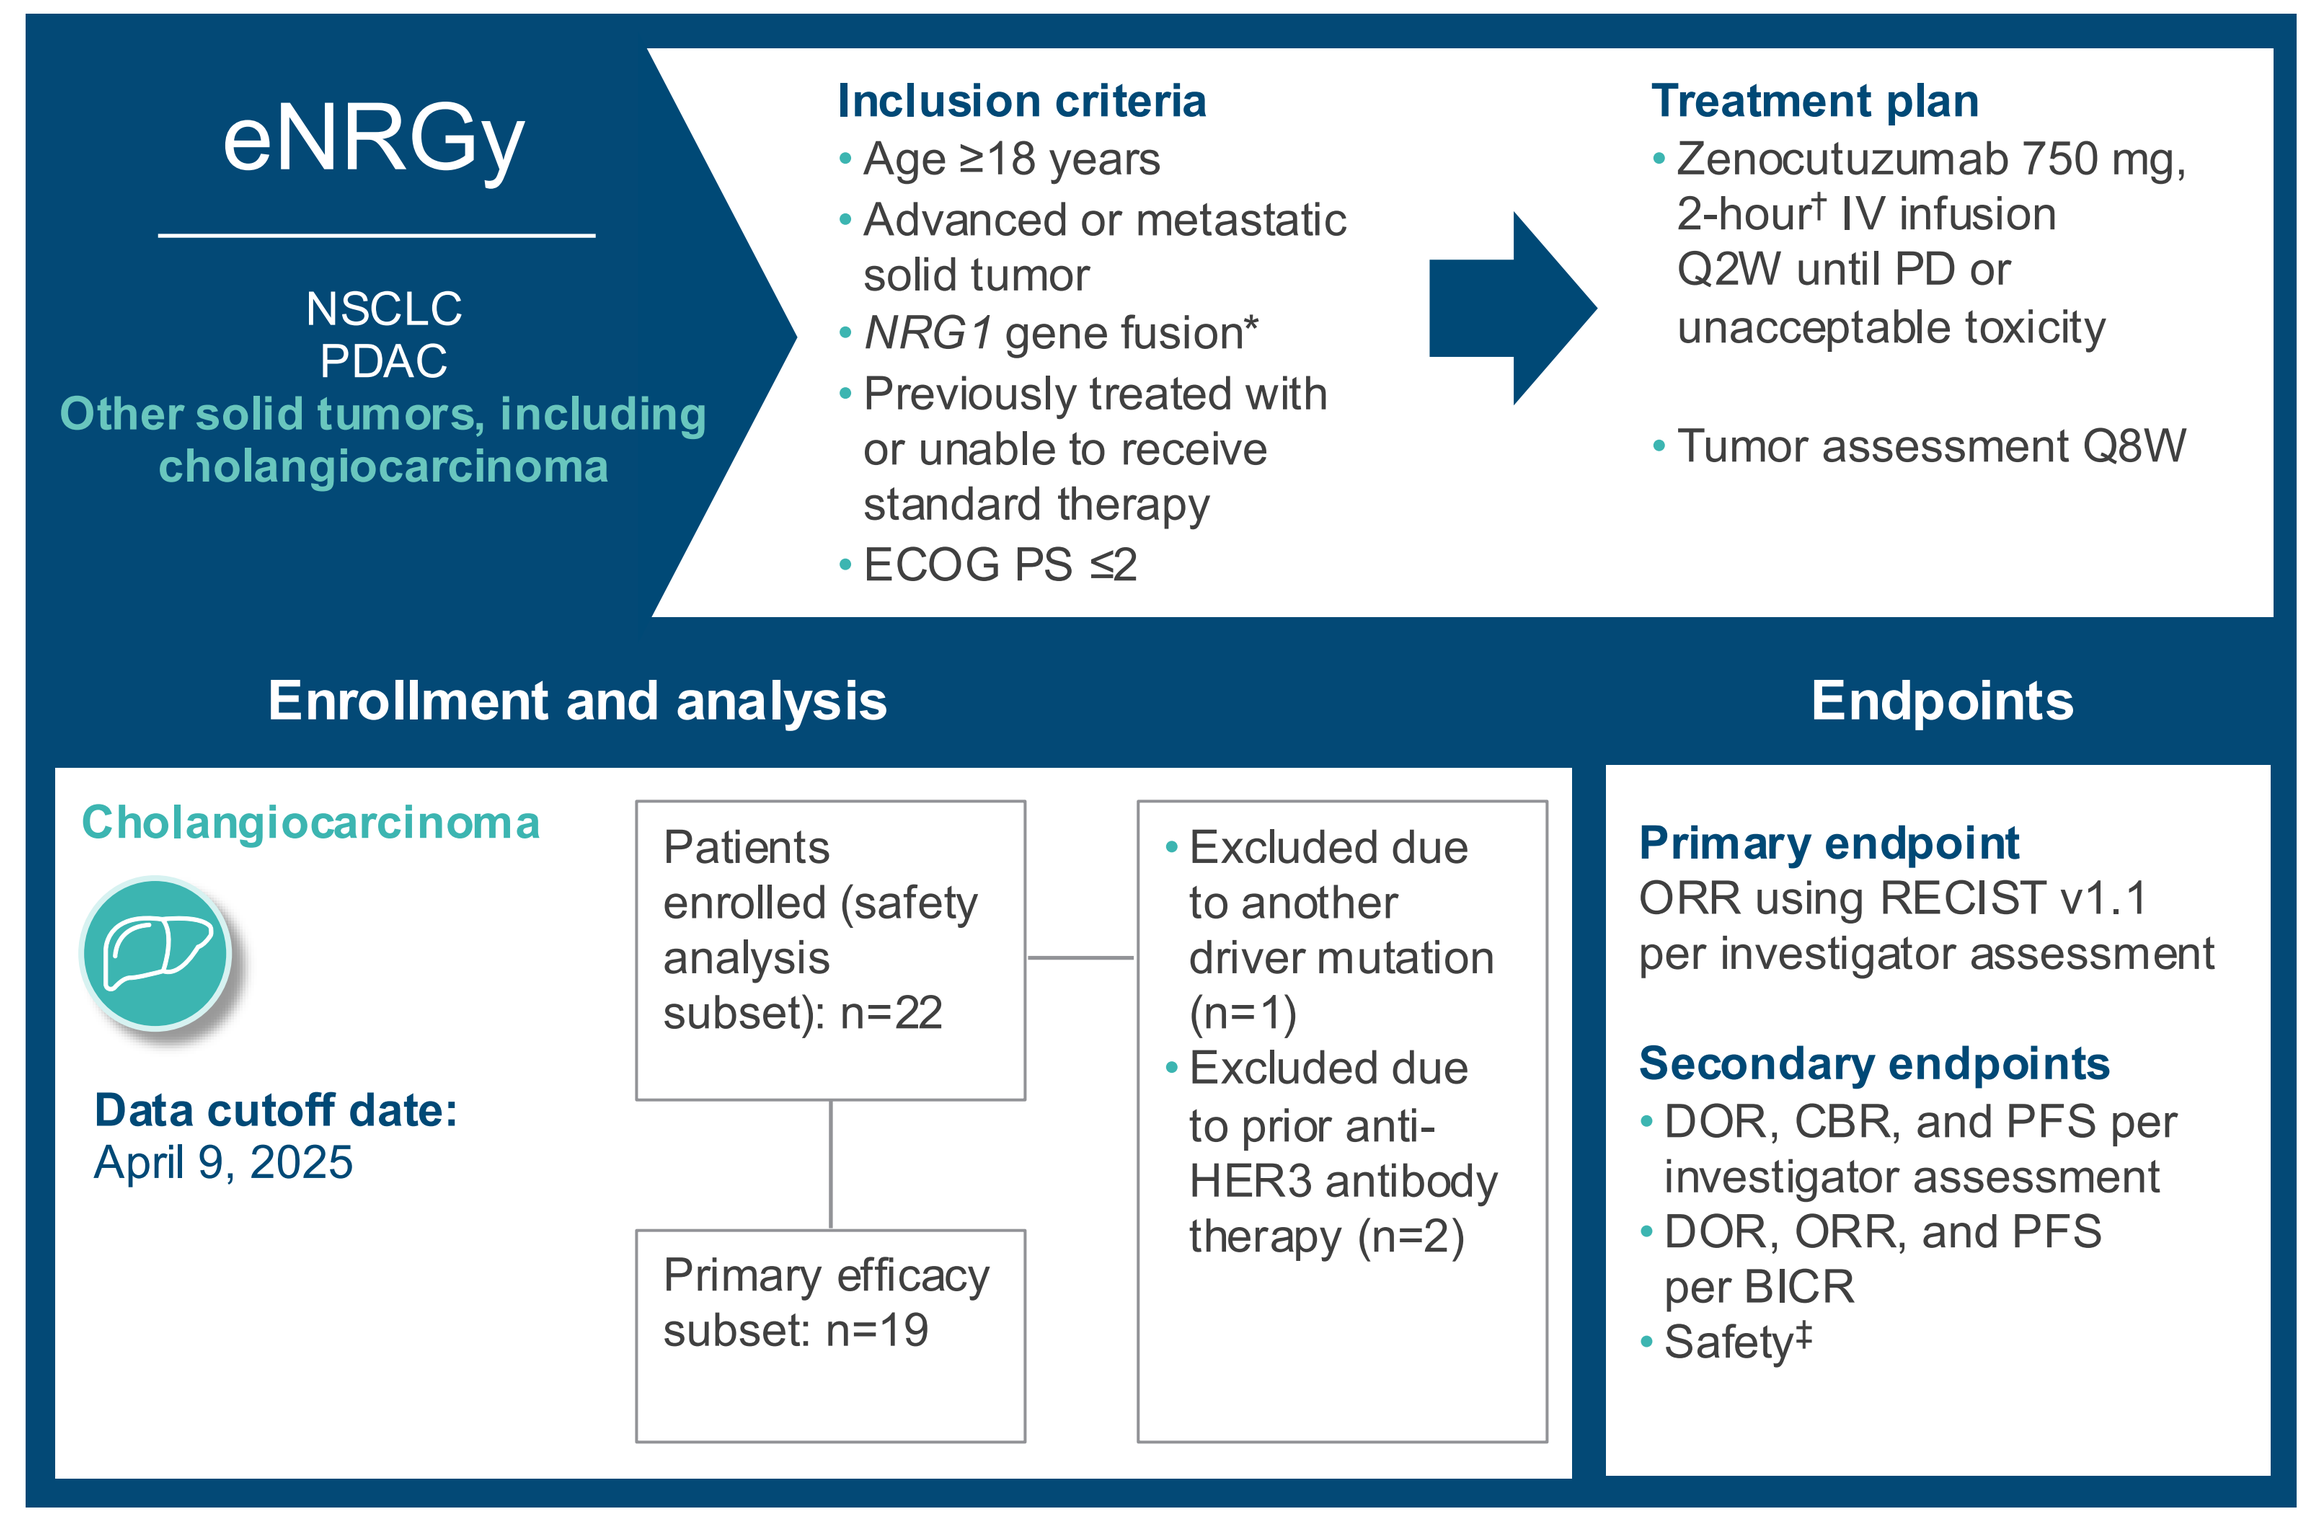
eNRGy
NSCLC
PDAC
Other solid tumors, including cholangiocarcinoma
Inclusion criteria
• Age ≥18 years
• Advanced or metastatic solid tumor
• *NRG1* gene fusion*
• Previously treated with or unable to receive standard therapy
• ECOG PS ≤2
Treatment plan
• Zenocutuzumab 750 mg, 2-hour† IV infusion Q2W until PD or unacceptable toxicity
• Tumor assessment Q8W
Enrollment and analysis
Cholangiocarcinoma
Data cutoff date: April 9, 2025
Patients enrolled (safety analysis subset): n=22
Primary efficacy subset: n=19
• Excluded due to another driver mutation (n=1)
• Excluded due to prior anti-HER3 antibody therapy (n=2)
Endpoints
Primary endpoint
ORR using RECIST v1.1 per investigator assessment
Secondary endpoints
• DOR, CBR, and PFS per investigator assessment
• DOR, ORR, and PFS per BICR
• Safety‡

*NRG1 gene fusion status was determined by NGS. †To mitigate potential IRRs, the initial infusion was administered over a period of 4 hours and patients received premedication with antipyretics, antihistamines, and glucocorticoids. ‡Adverse events were assessed from the date of the first zenocutuzumab dose up to 30 days after the last dose and graded using CTCAE v4.03.
BICR, blinded independent central review; CBR, clinical benefit rate; CTCAE, Common Terminology Criteria for Adverse Events; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; HER, human epidermal growth factor receptor; IRR, infusion-related reaction; IV, intravenous; NGS, next-generation sequencing; *NRG1*, neuregulin 1; NSCLC, non-small cell lung cancer; ORR, overall response rate; PD, progressive disease; PDAC, pancreatic adenocarcinoma; PFS, progression-free survival; Q2W, every 2 weeks; Q8W, every 8 weeks; RECIST, Response Evaluation Criteria in Solid Tumors.

Results

Table 1 | Demographics

Characteristic	Safety analysis set (N=22)
Age, years, median (range)	57.5 (23–82)
Male, n (%)	10 (45)
ECOG PS, n (%)	
0	14 (63)
1	7 (32)
2	1 (5)
Anatomical location, n (%)	
Intrahepatic	18 (82)
Unknown	4 (18)
Stage at screening, n (%)	
IIIB	1 (5)
IV	21 (95)

ECOG PS, Eastern Cooperative Oncology Group performance status.

Table 2 | Patient disposition

Disposition	Safety analysis set (N=22)
Study treatment disposition, n (%)	
Discontinued treatment	18 (82)
Treatment ongoing	4* (18)
Reason for discontinuation, n (%)	
Death	0 (0)
Lost to follow-up	1 (5)
Physician decision	1 (5)
Progressive disease	15 (68)
Withdrawal of consent	1 (5)

*Including 1 patient who is not in the primary efficacy set due to prior seribantumab.

Table 3 | Treatment history

Diagnosis and prior therapy	Primary efficacy set (N=19)
Time since metastatic diagnosis, months, median (range)	9.3 (1.6–34.2)
Patients receiving prior systemic therapy, n (%)	17 (89)
Type of prior therapy, n (%)	
Chemotherapy	16 (84)
Immunotherapy	3 (16)
Anti-VEGF therapy	1 (5)
Transarterial chemoembolization	1 (5)

VEGF, vascular endothelial growth factor.

Table 4 | Number of prior therapy regimens

Number of prior therapy regimens	Primary efficacy set (N=19)
Prior systemic therapy regimens, n, median (range)	1 (0–4)
Prior systemic therapy regimens, n (%)	
0	2 (11)
1	9 (47)
2	5 (26)
3	1 (5)
4	2 (11)

Figure 3 | Prior treatment exposure (left) and study treatment exposure and response (right) (primary efficacy set, n=19)

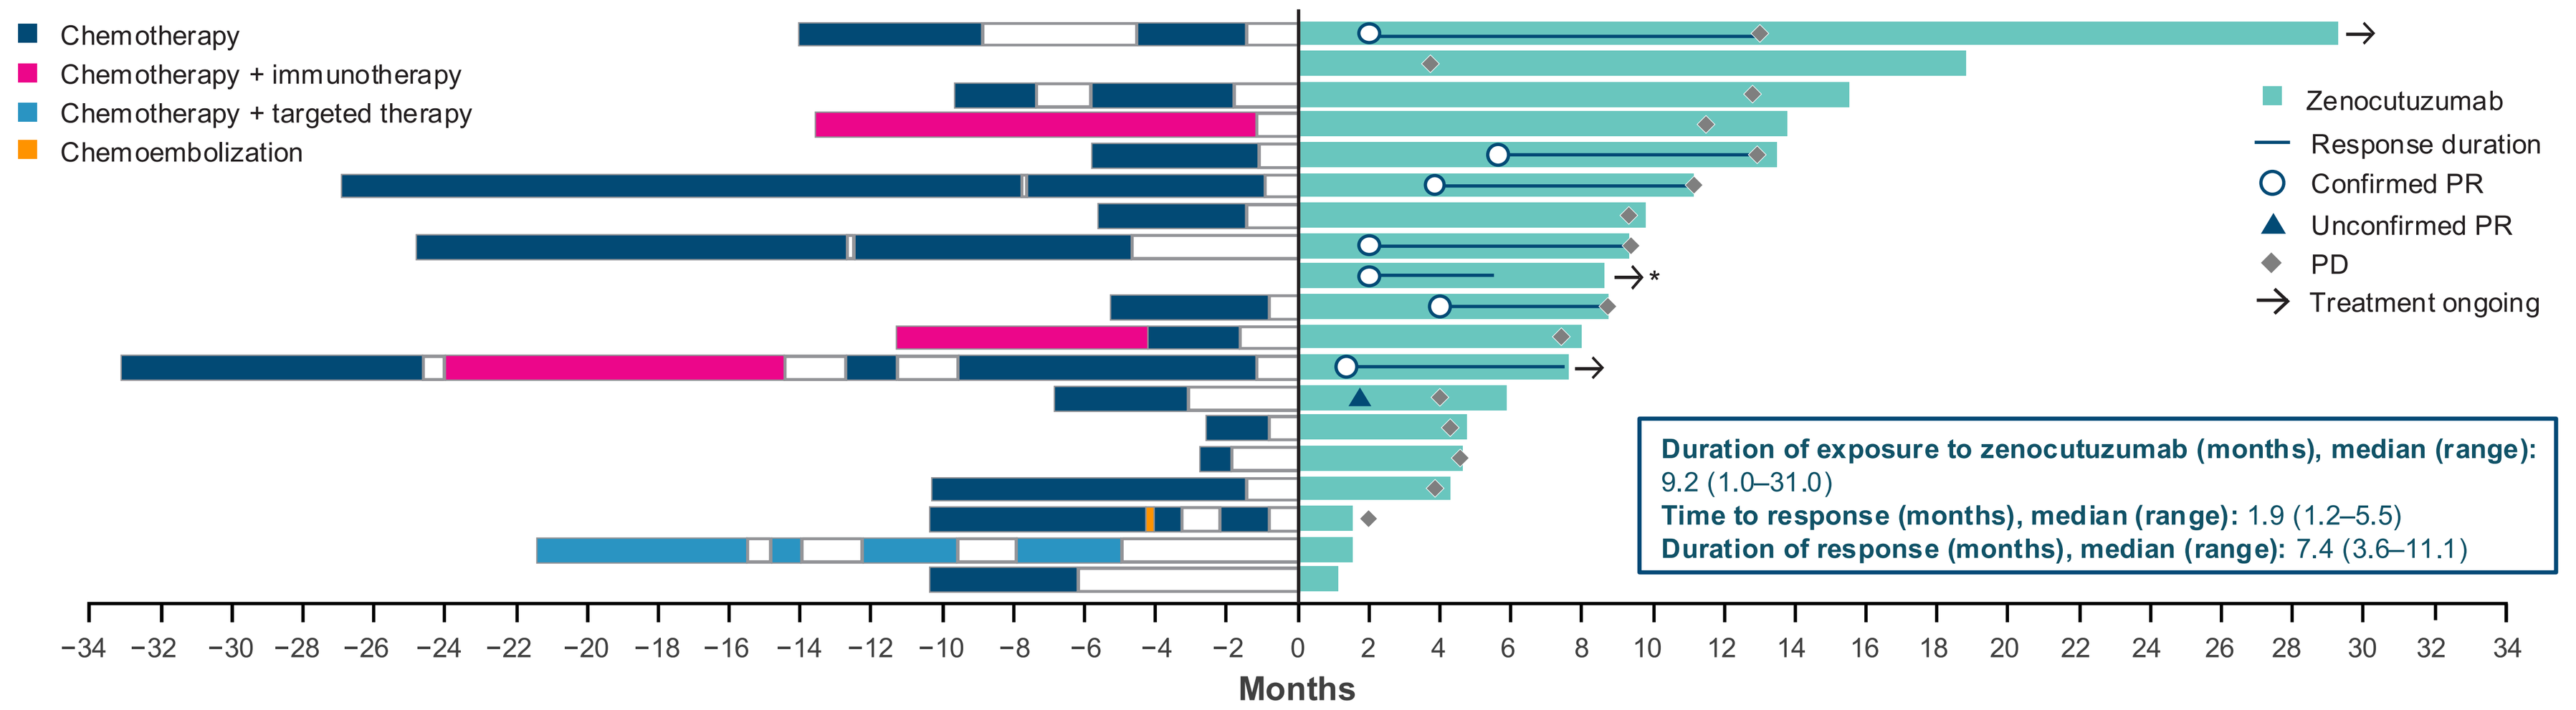
Chemotherapy
Chemotherapy + immunotherapy
Chemotherapy + targeted therapy
Chemoembolization
Zenocutuzumab
Response duration
Confirmed PR
Unconfirmed PR
PD
Treatment ongoing
Duration of exposure to zenocutuzumab (months), median (range): 9.2 (1.0–31.0)
Time to response (months), median (range): 1.9 (1.2–5.5)
Duration of response (months), median (range): 7.4 (3.6–11.1)
Months

*Response data missing due to data entry error; however, continued PR has been confirmed with the investigator.
PD, progressive disease; PR, partial response.

Figure 4 | Best overall response (primary efficacy set, n=19)

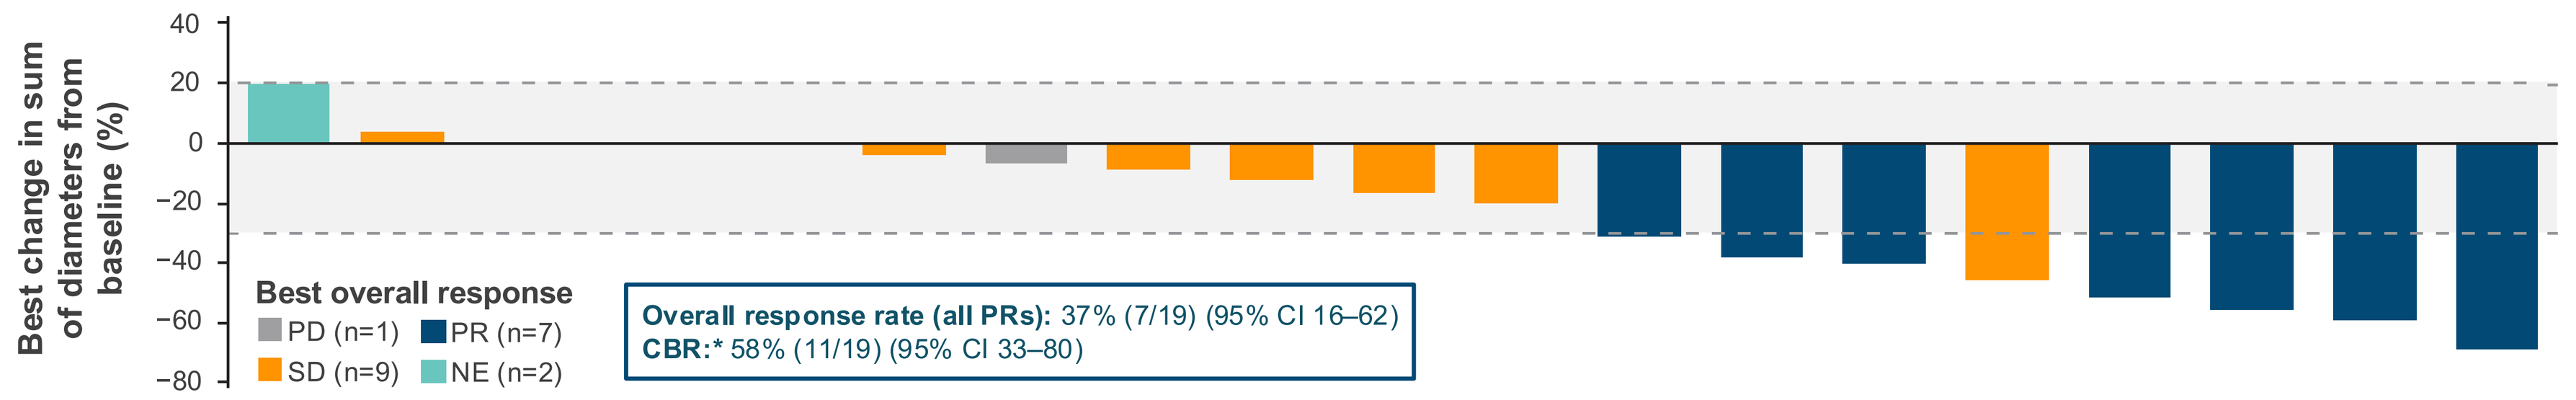
Best change in sum of diameters from baseline (%)
Best overall response
PD (n=1)
PR (n=7)
SD (n=9)
NE (n=2)
Overall response rate (all PRs): 37% (7/19) (95% CI 16–62)
CBR:* 58% (11/19) (95% CI 33–80)

*Defined as the proportion of patients who experienced a CR or PR or who had SD for ≥24 weeks.
CBR, clinical benefit rate; CI, confidence interval; CR, complete response; NE, not evaluable; PD, progressive disease; PR, partial response; SD, stable disease.

Figure 5 | Kaplan–Meier of PFS (primary efficacy set, n=19)

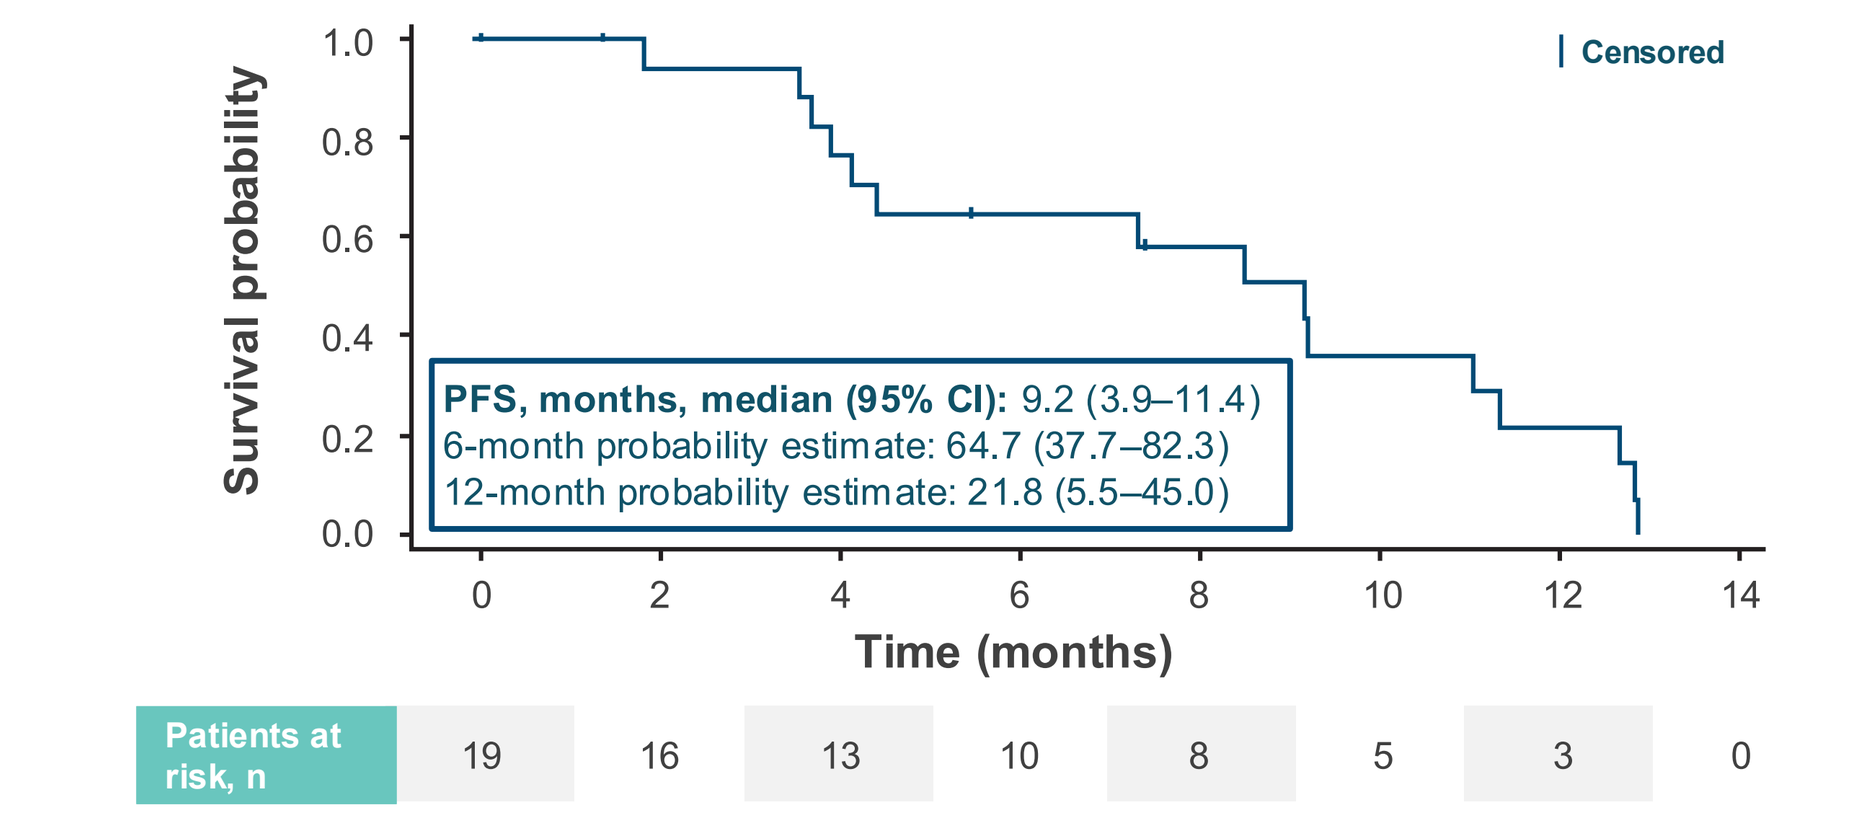
Survival probability
Censored
PFS, months, median (95% CI): 9.2 (3.9–11.4)
6-month probability estimate: 64.7 (37.7–82.3)
12-month probability estimate: 21.8 (5.5–45.0)
Time (months)
Patients at risk, n
19 16 13 10 8 5 3 0

CI, confidence interval; PFS, progression-free survival.

Figure 6 | Kaplan–Meier of OS (primary efficacy set, n=19)

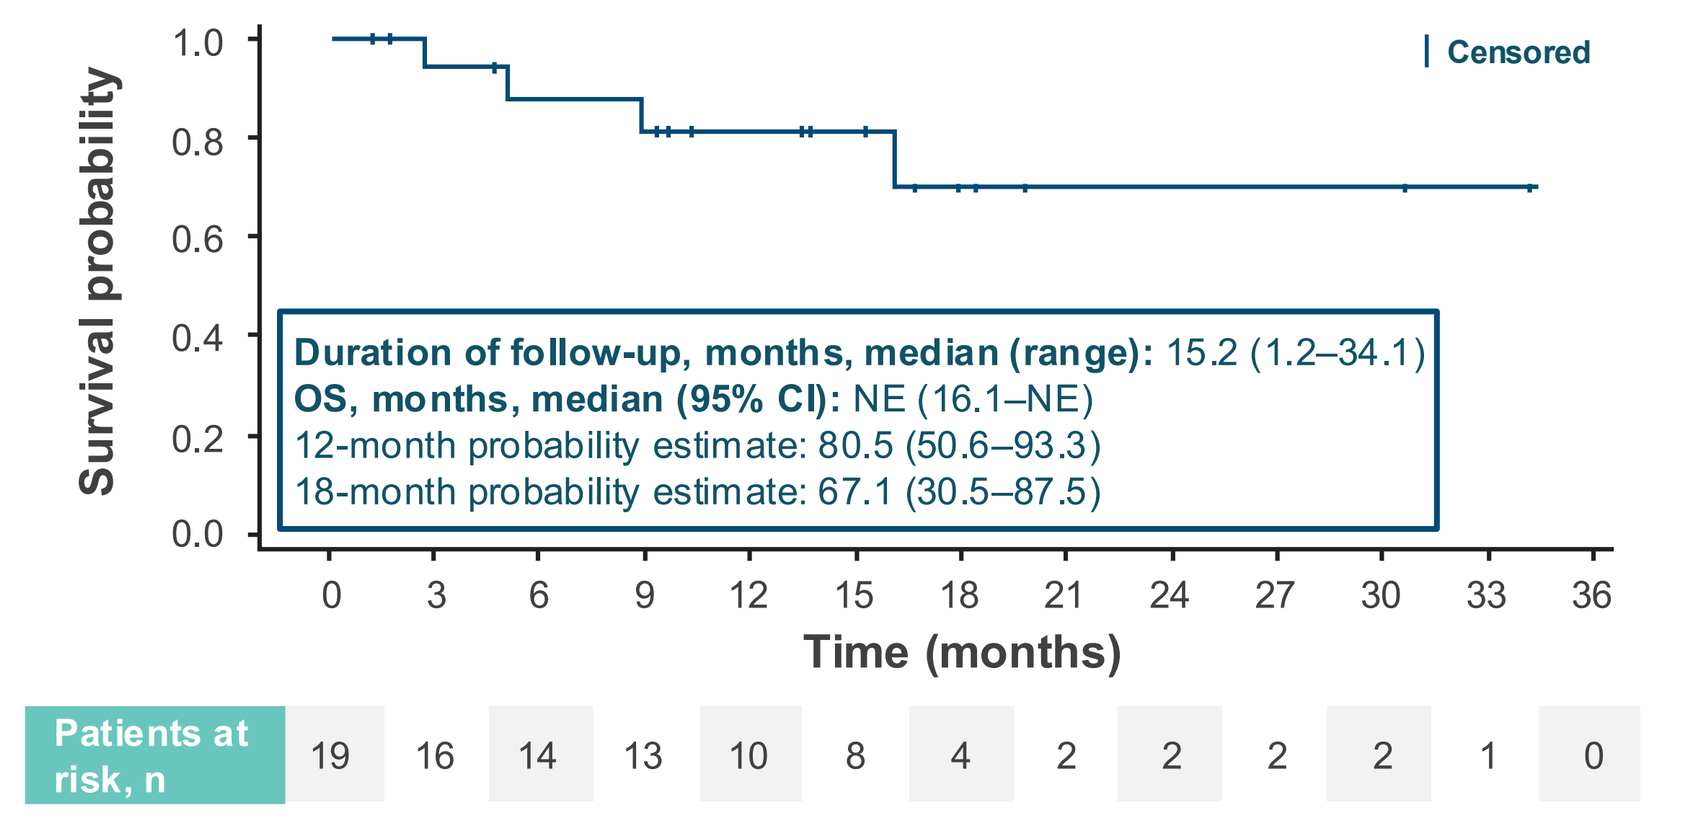
Survival probability
Censored
Duration of follow-up, months, median (range): 15.2 (1.2–34.1)
OS, months, median (95% CI): NE (16.1–NE)
12-month probability estimate: 80.5 (50.6–93.3)
18-month probability estimate: 67.1 (30.5–87.5)
Time (months)
Patients at risk, n
19 16 14 13 10 8 4 2 2 2 2 1 0

CI, confidence interval; NE, not estimable; OS, overall survival.

Figure 7 | Percent change from baseline in CA19-9 (A) and CEA (B) over time in biomarker-evaluable patients*

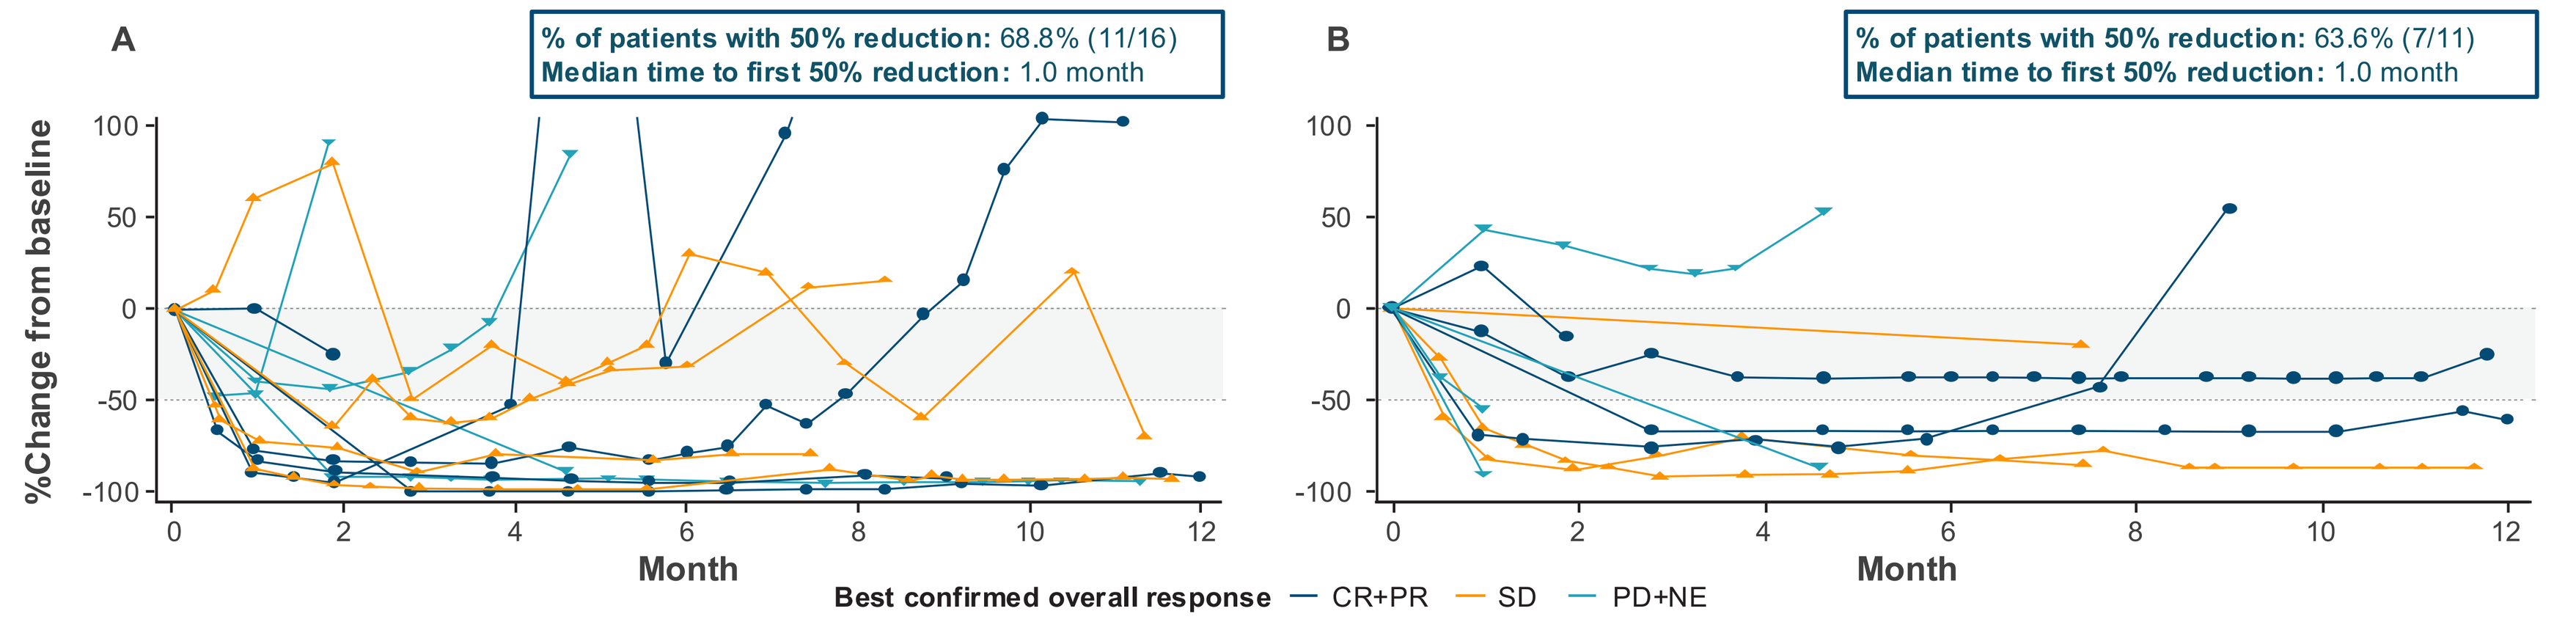
A
% of patients with 50% reduction: 68.8% (11/16)
Median time to first 50% reduction: 1.0 month
B
% of patients with 50% reduction: 63.6% (7/11)
Median time to first 50% reduction: 1.0 month
%Change from baseline
Month
Best confirmed overall response
CR+PR
SD
PD+NE

*Biomarker-evaluable patients defined as those with baseline and post-baseline measurements of CA19-9 (n=16) and CEA (n=11). Data points that go above the axis of these figures are censored.
CA19-9, carbohydrate antigen 19-9; CEA, carcinoembryonic antigen; CR, complete response; NE, not evaluable; PD, progressive disease; PR, partial response; SD, stable disease.

- CA19-9 declined in 100% of CA19-9–evaluable patients (16/16) including >50% reduction in 69% (11/16)
- Observed reductions in biochemical markers of disease provide additional evidence of the antitumor activity of zenocutuzumab

Table 5 | Safety summary

Adverse events	Safety analysis set, n (%) (N=22)	
Patients with at least one TEAE	21 (95)	
Patients with at least one TRAE	13 (59)	
Patients with at least one serious TEAE	5* (23)	
	All grades	Grade 3–4
Infusion-related reactions	2 (9)	0 (0)

*None were considered related to study drug. All were Grade ≤3 in severity.
TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

- No Grade 5 treatment-emergent adverse events (TEAEs) were reported
- A Grade ≥3 treatment-related AE (TRAE) was reported in 1 patient (5%, anemia)
 - No TRAEs led to study discontinuation
- Infusion-related reactions were reported in 2 patients (9%), both of which were Grade 1 or 2

Table 6 | TEAEs occurring in ≥20% of patients

Adverse events	Safety analysis set, n (%) (N=22)	
TEAEs occurring in ≥20% of patients	All grades	Grade 3–4
Anemia	10 (45)	3 (14)
Diarrhea	9 (41)	0 (0)
Hypomagnesemia	6 (27)	2 (9)
Abdominal pain	6 (27)	1 (5)
Cough	6 (27)	0 (0)
Fatigue	6 (27)	0 (0)
Nausea	6 (27)	0 (0)
ALT increased	5 (23)	1 (5)
GGT increased	2 (9)*	2 (9)

*GGT increase occurred in <20% of patients; however, they are included here as both events were Grade ≥3. Neither event was treatment-related. ALT, alanine aminotransferase; GGT, gamma-glutamyltransferase; TEAE, treatment-emergent adverse event.

CONCLUSIONS

Zenocutuzumab 750 mg Q2W demonstrated meaningful clinical activity in patients with *NRG1+* cholangiocarcinoma

- Overall response rate of 37% and clinical benefit rate (CBR) of 58%
 - CBR defined as the proportion of patients who had a complete or partial response, or had stable disease for ≥24 weeks
- Median time to response of 1.9 months; median duration of response of 7.4 months
- Median progression-free survival was 9.2 months; median overall survival has not been reached
- CA19-9 declined in 100% of CA19-9–evaluable patients (16/16) including >50% reduction in 69% (11/16)
- All patients with available subtype data (18/18) had intrahepatic cholangiocarcinoma

A manageable safety profile was observed with zenocutuzumab in this cohort

- Most TEAEs were Grade 1 or 2 and were manageable
- TRAEs occurred in 13 patients (59%); none led to study discontinuation
- No patients experienced a Grade 5 TEAE; no new safety concerns were identified

Results are consistent with those observed with zenocutuzumab in patients with other *NRG1+* solid tumors, including NSCLC and PDAC[9]

References
1. Banales JM, *et al. Nat Rev Gastroenterol Hepatol*. 2020;17(9):557–588; 2. Valle J, *et al. N Engl J Med*. 2010;362(14):1273–1281; 3. Oh DY, *et al. NEJM Evid*. 2022;1(8):EVIDoa2200015; 4. Kelley RK, *et al. Lancet*. 2023;401(10391):1853–1865; 5. Severson E, *et al. J Mol Diagn*. 2023;25(7):454–466; 6. Laskin J, *et al. Ann Oncol*. 2020;31(12):1693–1703; 7. Jonna S, *et al. Clin Cancer Res*. 2019;25(16):4966–4972; 8. US FDA. FDA grants accelerated approval to zenocutuzumab-zbco for non-small cell lung cancer and pancreatic adenocarcinoma. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-grants-accelerated-approval-zenocutuzumab-zbco-non-small-cell-lung-cancer-and-pancreatic. Accessed September 12, 2025; 9. Schram AM, *et al. N Engl J Med*. 2025;392(6):566–576; 10. ClinicalTrials.gov. NCT02912949. https://clinicaltrials.gov/study/NCT02912949. Accessed September 12, 2025.

Disclosures
Study sponsored by Merus N.V. and Partner Therapeutics, Inc. Alison M Schram has received advisory board compensation from Day One Biopharmaceuticals, Endeavor Biotherapeutics, Mersana, Merus, PMV Pharma, Relay Therapeutics, Repare, Revolution Medicines, and Schrödinger; received consultant fees from Blueprint Bio, Flagship Pioneering, Pro-Clin. Solutions LLC, and Redona Therapeutics; and has research funding paid to her institution from AstraZeneca, ArQule, BeiGene/SpringWorks, Black Diamond Therapeutics, Boehringer Ingelheim, Elevation Oncology, Kura, Lilly, Merus, Northern Biologics, Pfizer, PMV Pharma, Relay Therapeutics, Repare Therapeutics, Revolution Medicines, and Surface Oncology. Alison M Schram would like to acknowledge support from the ASCO Conquer Cancer Foundation CDA, NCI P30CA008748 CCITLA, Cycle for Survival, and Memorial Sloan Kettering Cancer Center Support Grant (P30 CA008748). Full author disclosures are available via the QR code.

Acknowledgments
We thank the patients and their families, the investigators and their teams, and Partner Therapeutics colleagues, clinical research organizations, and vendors. We gratefully acknowledge Merus N.V. for their contributions to lead the discovery, development and successful approval of zenocutuzumab. Medical writing support was provided by Eleanor Steele and Emmaline Tregembo of LiNK Health Group Ltd.

CONGRESS INFORMATION: AACR-NCI-EORTC Molecular Targets and Cancer Therapeutics; Boston, MA, USA; October 22–26, 2025

CONTACT INFORMATION: Alison M Schram, Memorial Sloan Kettering Cancer Center, New York, NY, USA; schrama@mskcc.org